The Effect of ASP6432, a Novel Type 1 Lysophosphatidic Acid Receptor Antagonist, on Urethral Function and Prostate Cell Proliferation

156

Sakamoto K[1], Noguchi Y[1], Ueshima K[1], Yamakuni H[2], Ohtake A[1], Sato S[1], Ishizu K[1], Hosogai N[1], Kawaminami E[1], Takeda M[1] and Masuda N[1]
1. Astellas Pharma Inc., 2. Astellas Research Technologies Co., Ltd

Background, Aim, Materials and Methods

- Pharmacotherapy for BPH still remains room for improvement.
- Lysophosphatidic acid (LPA) is a bioactive phospholipid reported to contract urethra[1] and proliferate prostate cells[2]. However, details including responsible receptor have not been fully elucidated.
- We explored the role of LPA and type 1 LPA receptor (LPA1R) on urethral and prostatic functions using ASP6432, a newly discovered LPA1R antagonist.
 - Antagonist assays using cells expressing human LPA1R to LPA5R
 - Contraction study in prostate and urethral strips isolated from rats
 - Urethral pressure measurement in anesthetized rats (urethane, 1.2 g/kg ip)
 - Cell proliferation assay [Bromodeoxyuridine (BrdU) incorporation] in primary cultured human prostate stroma cells (PrSC)

[1]Saga et al., PLoS One. 2014;9(4):e93230. [2]Adolfsson et al., Prostate. 2002;51(1):50-8.

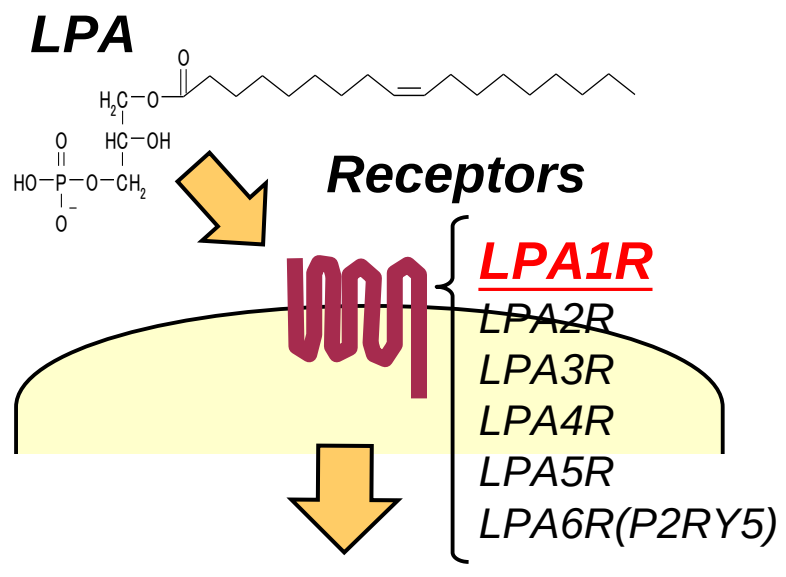
LPA
Receptors
LPA1R
LPA2R
LPA3R
LPA4R
LPA5R
LPA6R(P2RY5)

- *Smooth muscle contraction*
- *Cell proliferation and survival*
- *Mechanical stress response enhancement*
- *Afferent nerve activation*

Results

ASP6432 exhibited a potent antagonistic activity on LPA1R with >10 times selectivity over other LPA receptor subtypes.

Effects of ASP6432 on LPA-induced increase in intracellular Ca^{2+} (LPA1R to 4R) or cyclic AMP (LPA5R)

Subtypes	LPA1R	LPA2R	LPA3R	LPA4R	LPA5R
IC_{50} value (nmol/L)	**11**	**> 10000**	**> 10000**	**113.9**	**>30000**

Mean of 2 to 4 independent experiments

ASP6432 concentration-dependently inhibited LPA (100 μmol/L)-induced contractions in rat prostate and urethral strips.

Prostate

Mean±SEM, N=5
∗ p<0.025 vs. Vehicle (Williams' multiple comparison test)

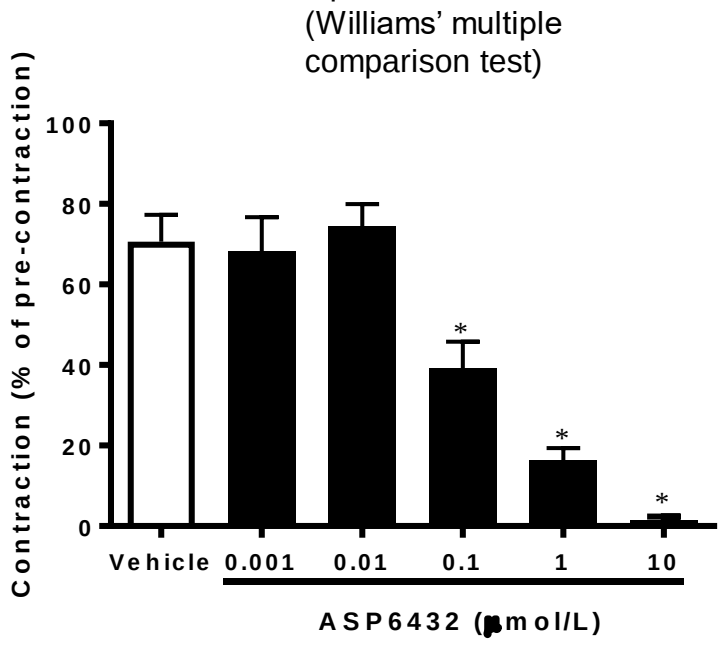
Contraction (% of pre-contraction)
100
80
60
40
20
0
Vehicle
0.001
0.01
0.1
1
10
ASP6432 (μmol/L)

Urethra

Mean±SEM, N=5
∗ p<0.025 vs. Pre (Williams' multiple comparison test using within subject error)

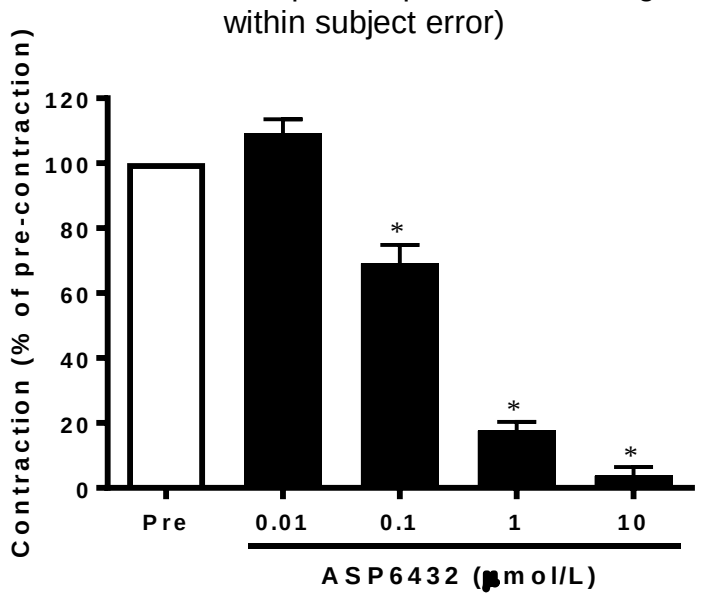
Contraction (% of pre-contraction)
120
100
80
60
40
20
0
Pre
0.01
0.1
1
10
ASP6432 (μmol/L)

ASP6432 dose-dependently inhibited LPA (3 mg/kg iv)-induced intraurethral pressure increase.

Mean±SEM, N=6
∗∗ p<0.01 vs. Vehicle (Dunnett's test using within subject error)

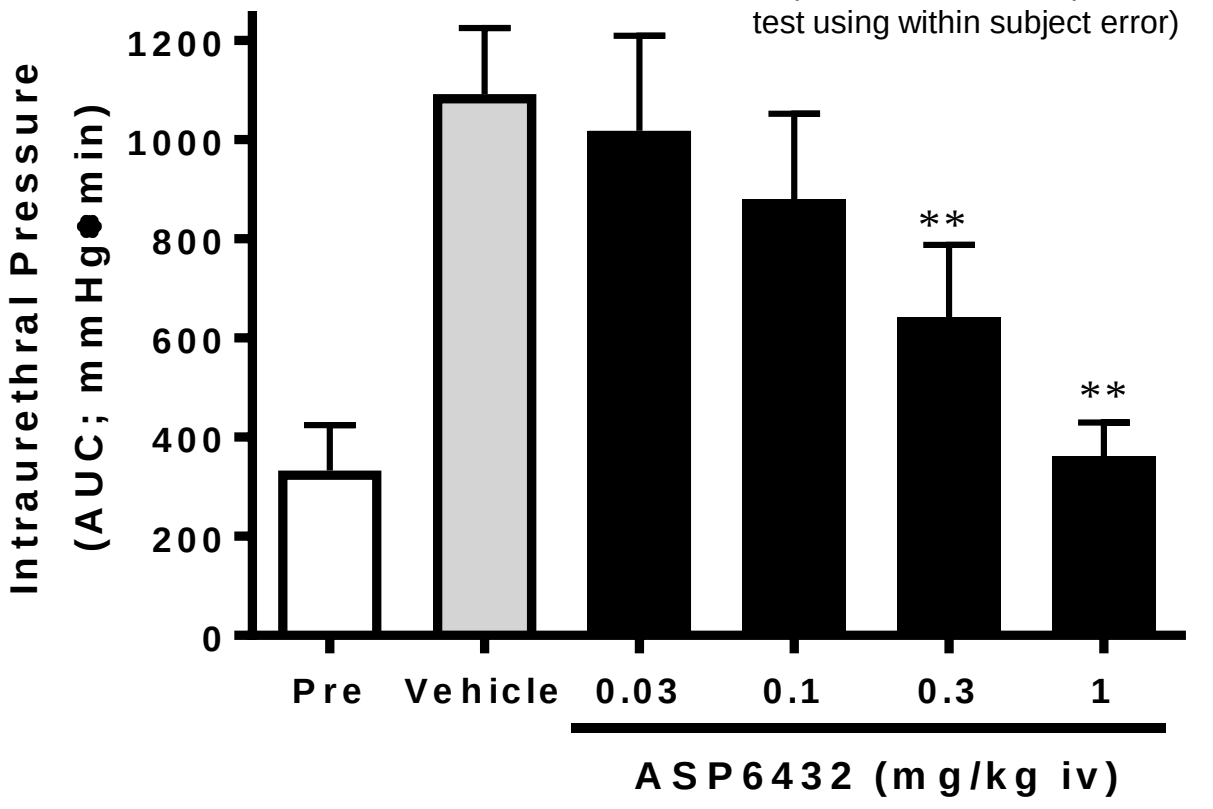
Intraurethral Pressure
(AUC; mmHg●min)
1200
1000
800
600
400
200
0
Pre
Vehicle
0.03
0.1
0.3
1
ASP6432 (mg/kg iv)

ASP6432 dose-dependently decreased urethral perfusion pressure (UPP) with a maximum reduction (42.5%) larger than that with a sufficient dose of tamsulosin[3] (Tam, 15.4% at 10 μg/kg iv), an α1 adrenoceptor antagonist.

[3]Akiyama et al., J Pharmacol Exp Ther. 1999;291(1):81-91.

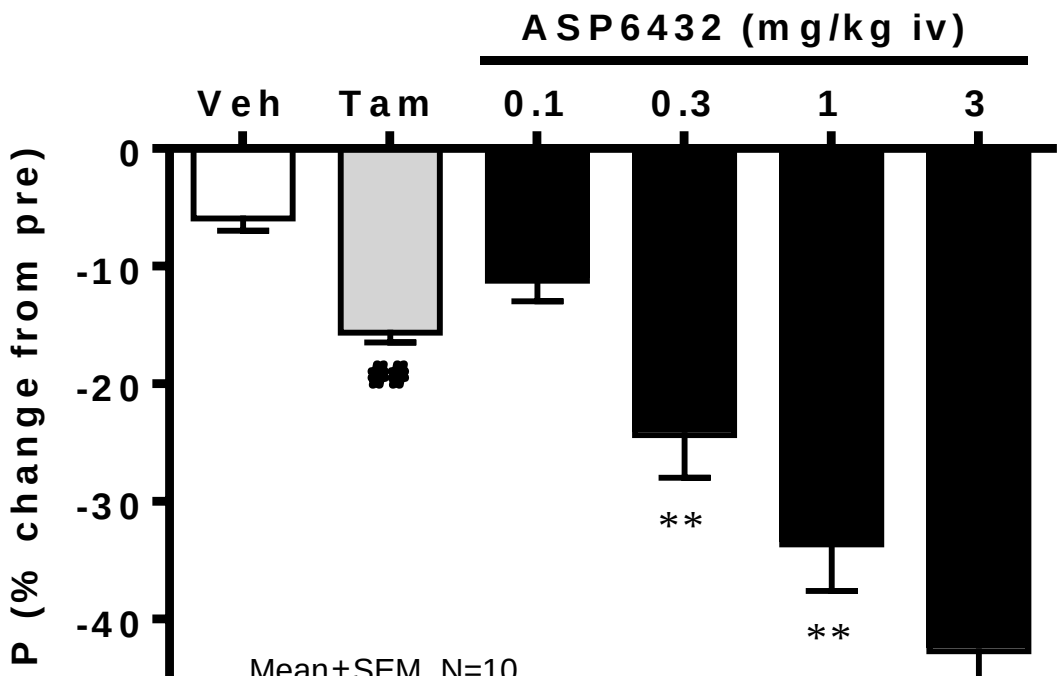
ASP6432 (mg/kg iv)
Veh
Tam
0.1
0.3
1
3
UPP (% change from pre)
0
-10
-20
-30
-40
-50
##
**
**
**

Mean±SEM, N=10
p<0.01 vs. Vehicle (Veh) (Student's t-test),
∗∗ P<0.01 vs. Vehicle (Dunnett's multiple comparison test)

ASP6432 suppressed LPA (10 μmol/L)-induced human prostate stromal cell proliferation.

Mean±SEM, N=4
p<0.05 vs. Normal (N) (Paired t-test),
∗ p<0.025 vs. Control (C) (William's multiple comparison test using within subject error)

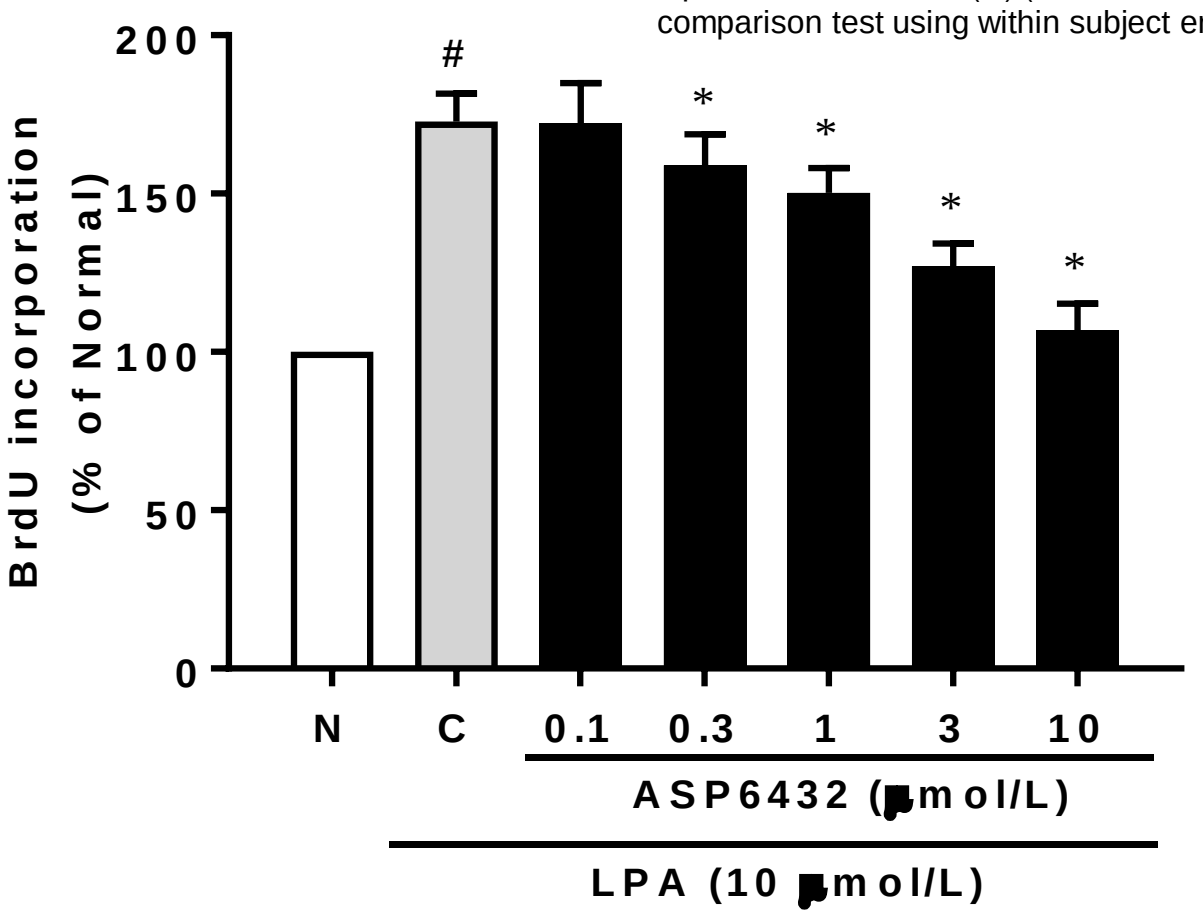
BrdU incorporation
(% of Normal)
200
150
100
50
0
#
N
C
0.1
0.3
1
3
10
ASP6432 (μmol/L)
LPA (10 μmol/L)

Discussions and Conclusions

- ASP6432 suppressed LPA-induced prostatic/urethral contractions and prostate cell proliferation, indicating the responsible role of LPA1R in these LPA-induced biological responses.
- ASP6432 decreased the urethral perfusion pressure (=baseline urethral pressure under no exogenous LPA stimulation) to a greater extent than that of tamsulosin, suggesting a significant role of LPA and LPA1R in the regulation of urethral tonus.
- With a potent urethral relaxation effect and its potential on prostatic stromal growth inhibition, ASP6432 could be a novel therapeutic agent for BPH.

Disclosure statement: all authors are employees of the Astellas group.